Weight Reduction After 6 Months of Setmelanotide Treatment in Patients With Hypothalamic Obesity

Christian L. Roth, MD[1,2]; Ashley H. Shoemaker, MD[3]; Michael Gottschalk, MD, PhD[4]; Jennifer Miller, MD, MS[5]; Guojun Yuan, PhD[6]; Sonali Malhotra, MD[6-8]; Cecilia Scimia, MD, PhD[6]; M. Jennifer Abuzzahab, MD[9]

[1]Seattle Children's Research Institute, Seattle, WA, USA; [2]Division of Endocrinology, Department of Pediatrics, University of Washington, Seattle, WA, USA; [3]Ian Burr Division of Endocrinology and Diabetes, Vanderbilt University Medical Center, Nashville, TN, USA; [4]Pediatric Endocrinology, University of California San Diego/Rady Children's Hospital, San Diego, CA, USA; [5]Pediatric Endocrinology, Department of Pediatrics, College of Medicine, University of Florida, Gainesville, FL, USA; [6]Rhythm Pharmaceuticals, Inc., Boston, MA, USA; [7]Massachusetts General Hospital, Boston, MA, USA; [8]Harvard Medical School, Boston, MA, USA; [9]Pediatric Endocrinology and Diabetes, Children's Minnesota, Saint Paul, MN, USA

SAT-656

Presenting Author:
Christian Roth
christian.roth@seattlechildrens.org

Summary

- **Six months of treatment with setmelanotide resulted in meaningful weight-related changes in a heterogenous population of patients with hypothalamic obesity (HO)**

Introduction

- HO is an acquired obesity and is characterized by rapid weight gain following insult to the hypothalamus[1-3]
 - Damage to the hypothalamus resulting from tumor invasion, radiotherapy, or surgical resection can impair leptin-melanocortin signaling, contributing to HO etiology[1-3]
- Patients with HO are generally refractory to standard treatment strategies for obesity management[1-2]
- In a Phase 2 trial of the melanocortin-4 receptor (MC4R) agonist setmelanotide, a synthetic α–melanocyte-stimulating hormone analogue, patients with HO experienced clinically meaningful and significant weight loss and reduction in hunger after 16 weeks of treatment[4]
 - All adherent patients (n=17) experienced weight loss with 16 weeks of setmelanotide treatment
 - Mean change in body mass index (BMI) at Week 16 was −14.9% (n=17)

Objective

- To report age-appropriate weight-related parameters after 6 months of treatment with setmelanotide

Methods

Trial design

- Patients aged 6 to 40 years from a Phase 2 multicenter, open-label study of setmelanotide (NCT04725240) were eligible to enroll in a long-term extension (LTE) trial (NCT03651765) if they experienced ≥5% reduction in BMI or clinically meaningful benefit as determined by a study investigator and exhibited adequate safety after 16 weeks of treatment
- During the index trial, patients underwent age-dependent dose titration over 4 weeks to a maximum dose of 3.0 mg of setmelanotide administered subcutaneously once daily for a total of 16 weeks of treatment
- Setmelanotide treatment throughout the LTE was at the same dose established during the index trial

Outcomes

- The proportion of patients achieving ≥5% and ≥10% reduction in BMI was assessed at Month 6 of treatment
- Additional outcomes assessed at Month 6 of treatment included
 - Mean percent change in body weight for patients ≥18 years old
 - Mean change in BMI Z score and percent of the 95th percentile for BMI (%BMI95) for patients <18 years old
 - Mean change in body composition across patients
 - Frequency of adverse events (AEs)

Results

Patient disposition and baseline characteristics

- Of 18 patients who enrolled in the index trial, 14 (77.8%) continued into the LTE and 13 (72.2%) had received ≥6 months of setmelanotide at the time of the analysis (Figure 1)

Figure 1. Patient disposition.

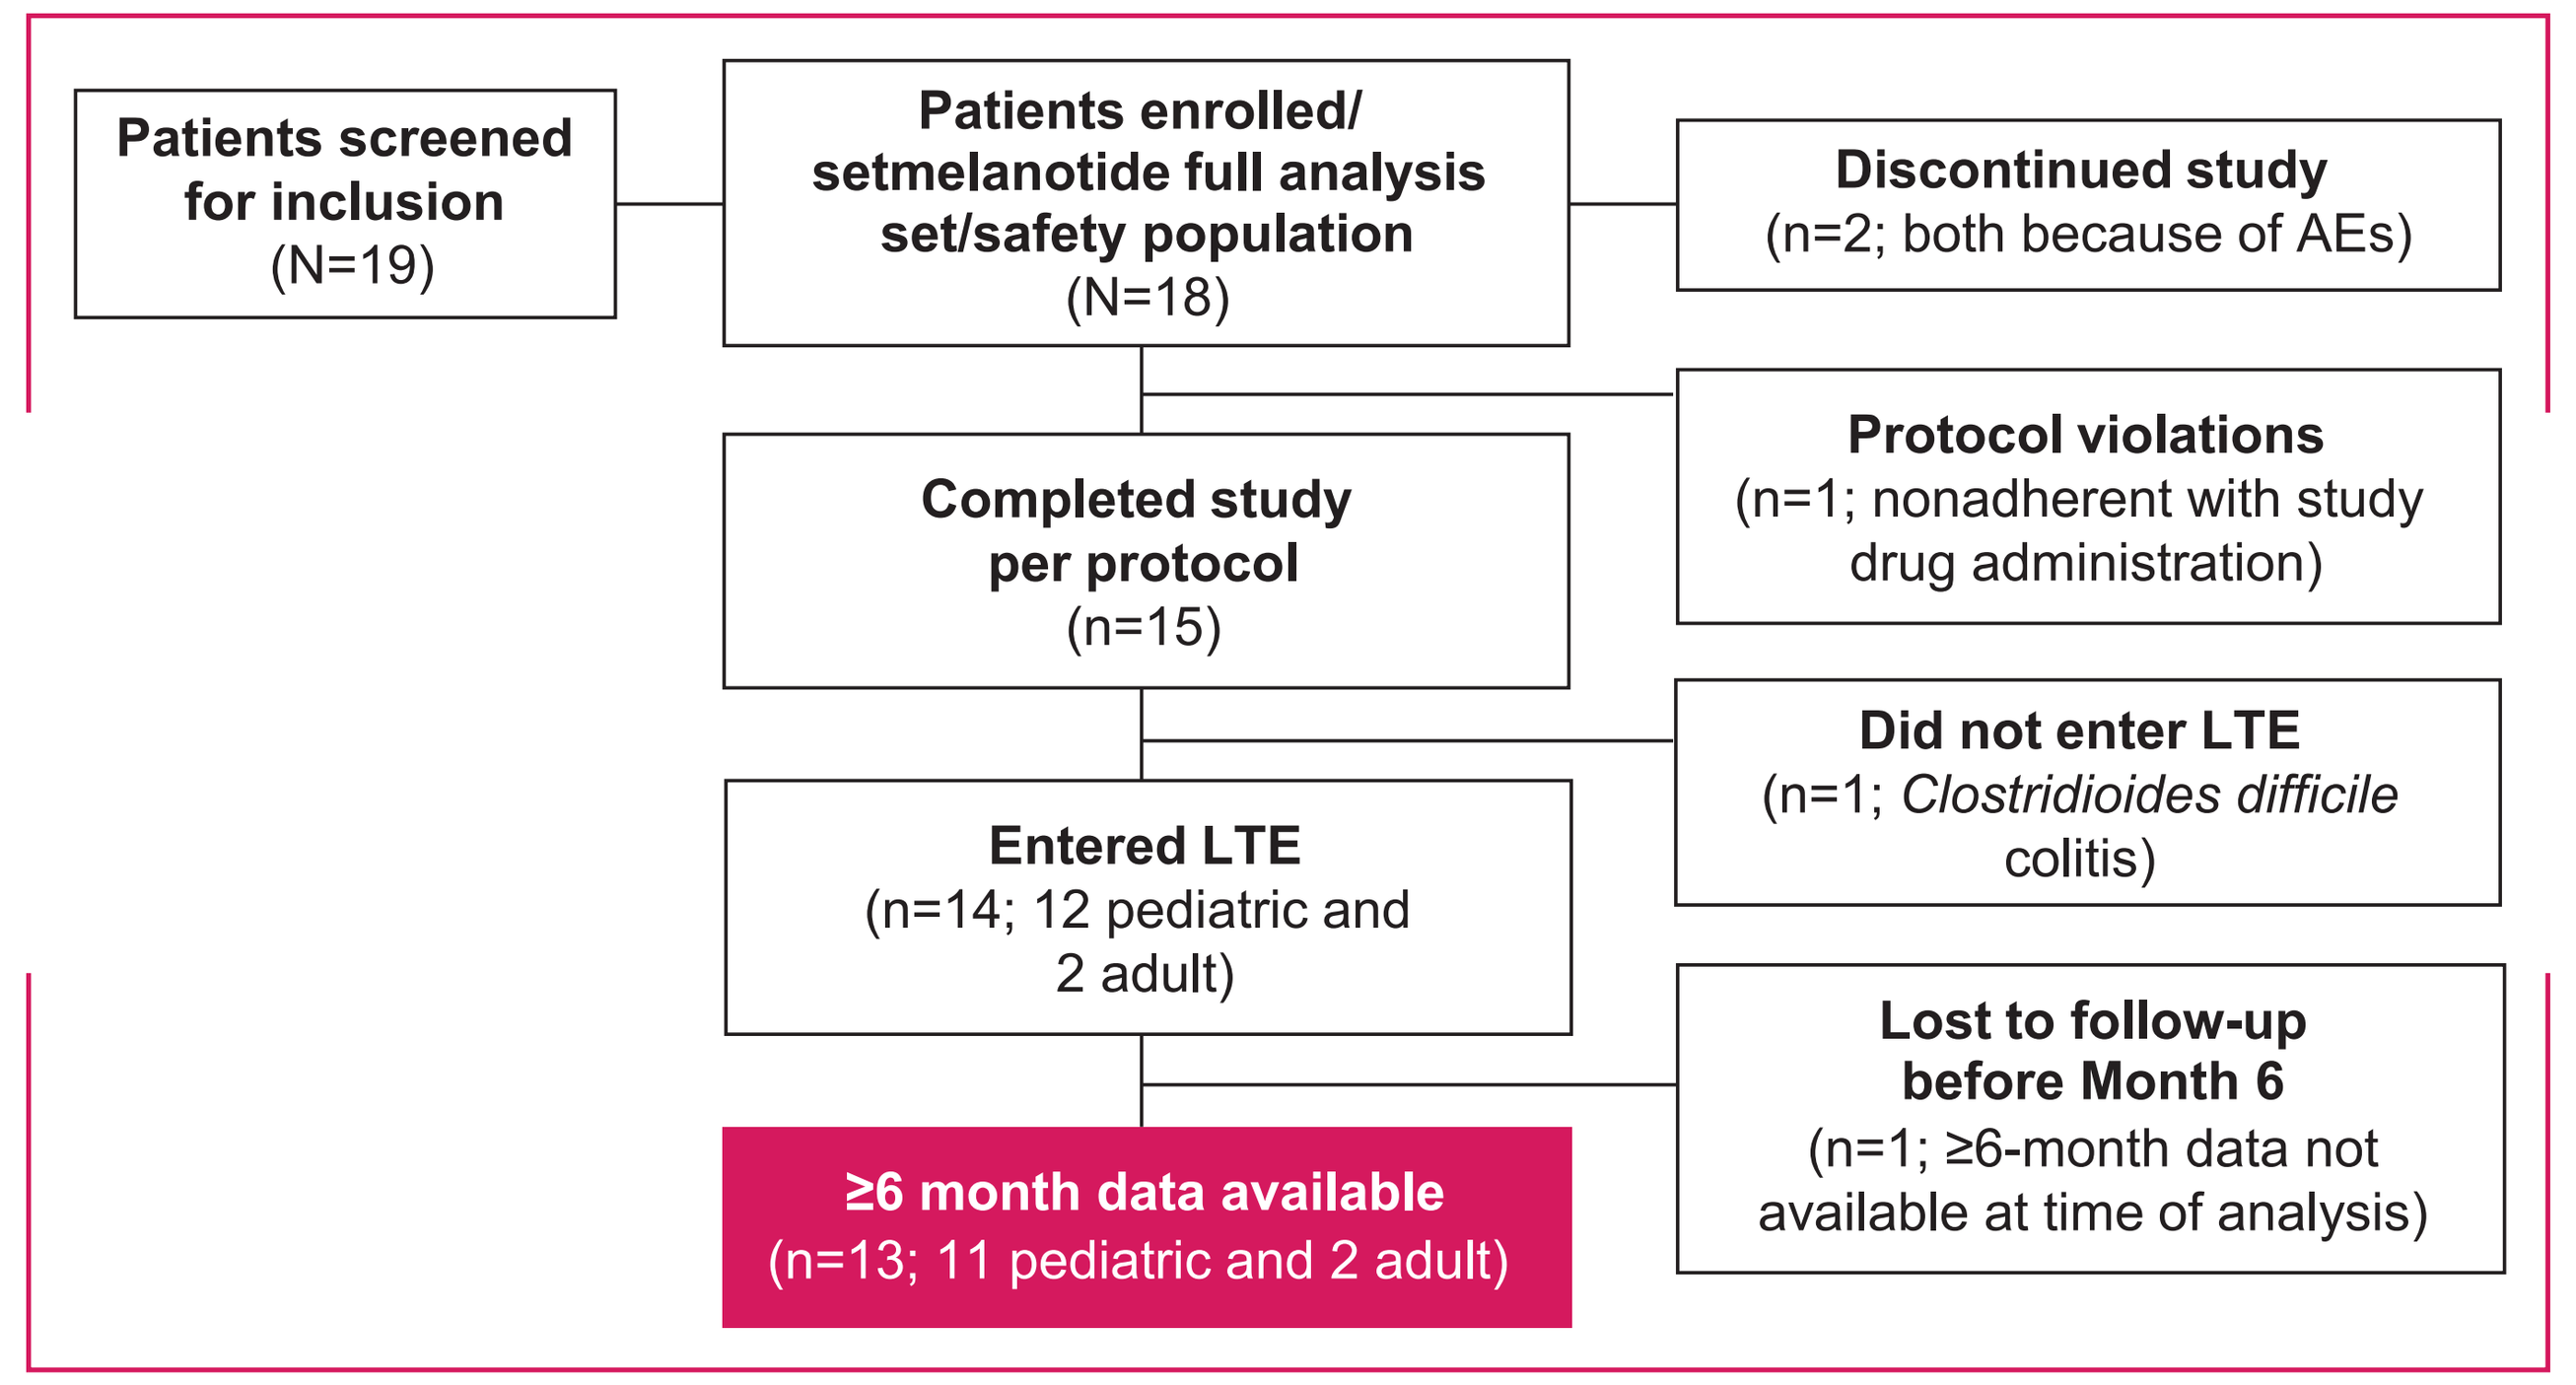
Patients screened for inclusion (N=19)
Patients enrolled/ setmelanotide full analysis set/safety population (N=18)
Discontinued study (n=2; both because of AEs)
Protocol violations (n=1; nonadherent with study drug administration)
Completed study per protocol (n=15)
Did not enter LTE (n=1; Clostridioides difficile colitis)
Entered LTE (n=14; 12 pediatric and 2 adult)
Lost to follow-up before Month 6 (n=1; ≥6-month data not available at time of analysis)
≥6 month data available (n=13; 11 pediatric and 2 adult)

AE, adverse event; LTE, long-term extension.

- Most patients in this analysis were <18 years of age at study entry, had treatment for craniopharyngioma, and were also being treated with multiple agents for complex comorbidities (Table 1)

Table 1. Demographics and Baseline Characteristics (n=13)

	Patients, n (%)
Age	
≥18 years	2 (15.4)
<18 years	11 (84.6)
Male	9 (69.2)
Race	
White	10 (76.9)
Black or African American	2 (15.4)
Native Hawaiian or Other Pacific Islander	1 (7.7)
Ethnicity	
Not Hispanic or Latino	10 (76.9)
Hispanic or Latino	3 (23.1)
Concomitant Medications reported in ≥50% of patients	
Hydrocortisone	11 (84.6)
Levothyroxine	9 (69.2)
Somatropin	9 (69.2)
Desmopressin Acetate	7 (53.8)
Ondansetron*	7 (53.8)
Tumor type	
Craniopharyngioma	10 (76.9)
Hypothalamic hamartoma	2 (15.3)
Juvenile pilocytic astrocytoma	1 (7.7)

*Ondansetron was taken for treatment of nausea and vomiting.

Efficacy outcomes

Figure 2. Individual BMI% change from baseline over time.

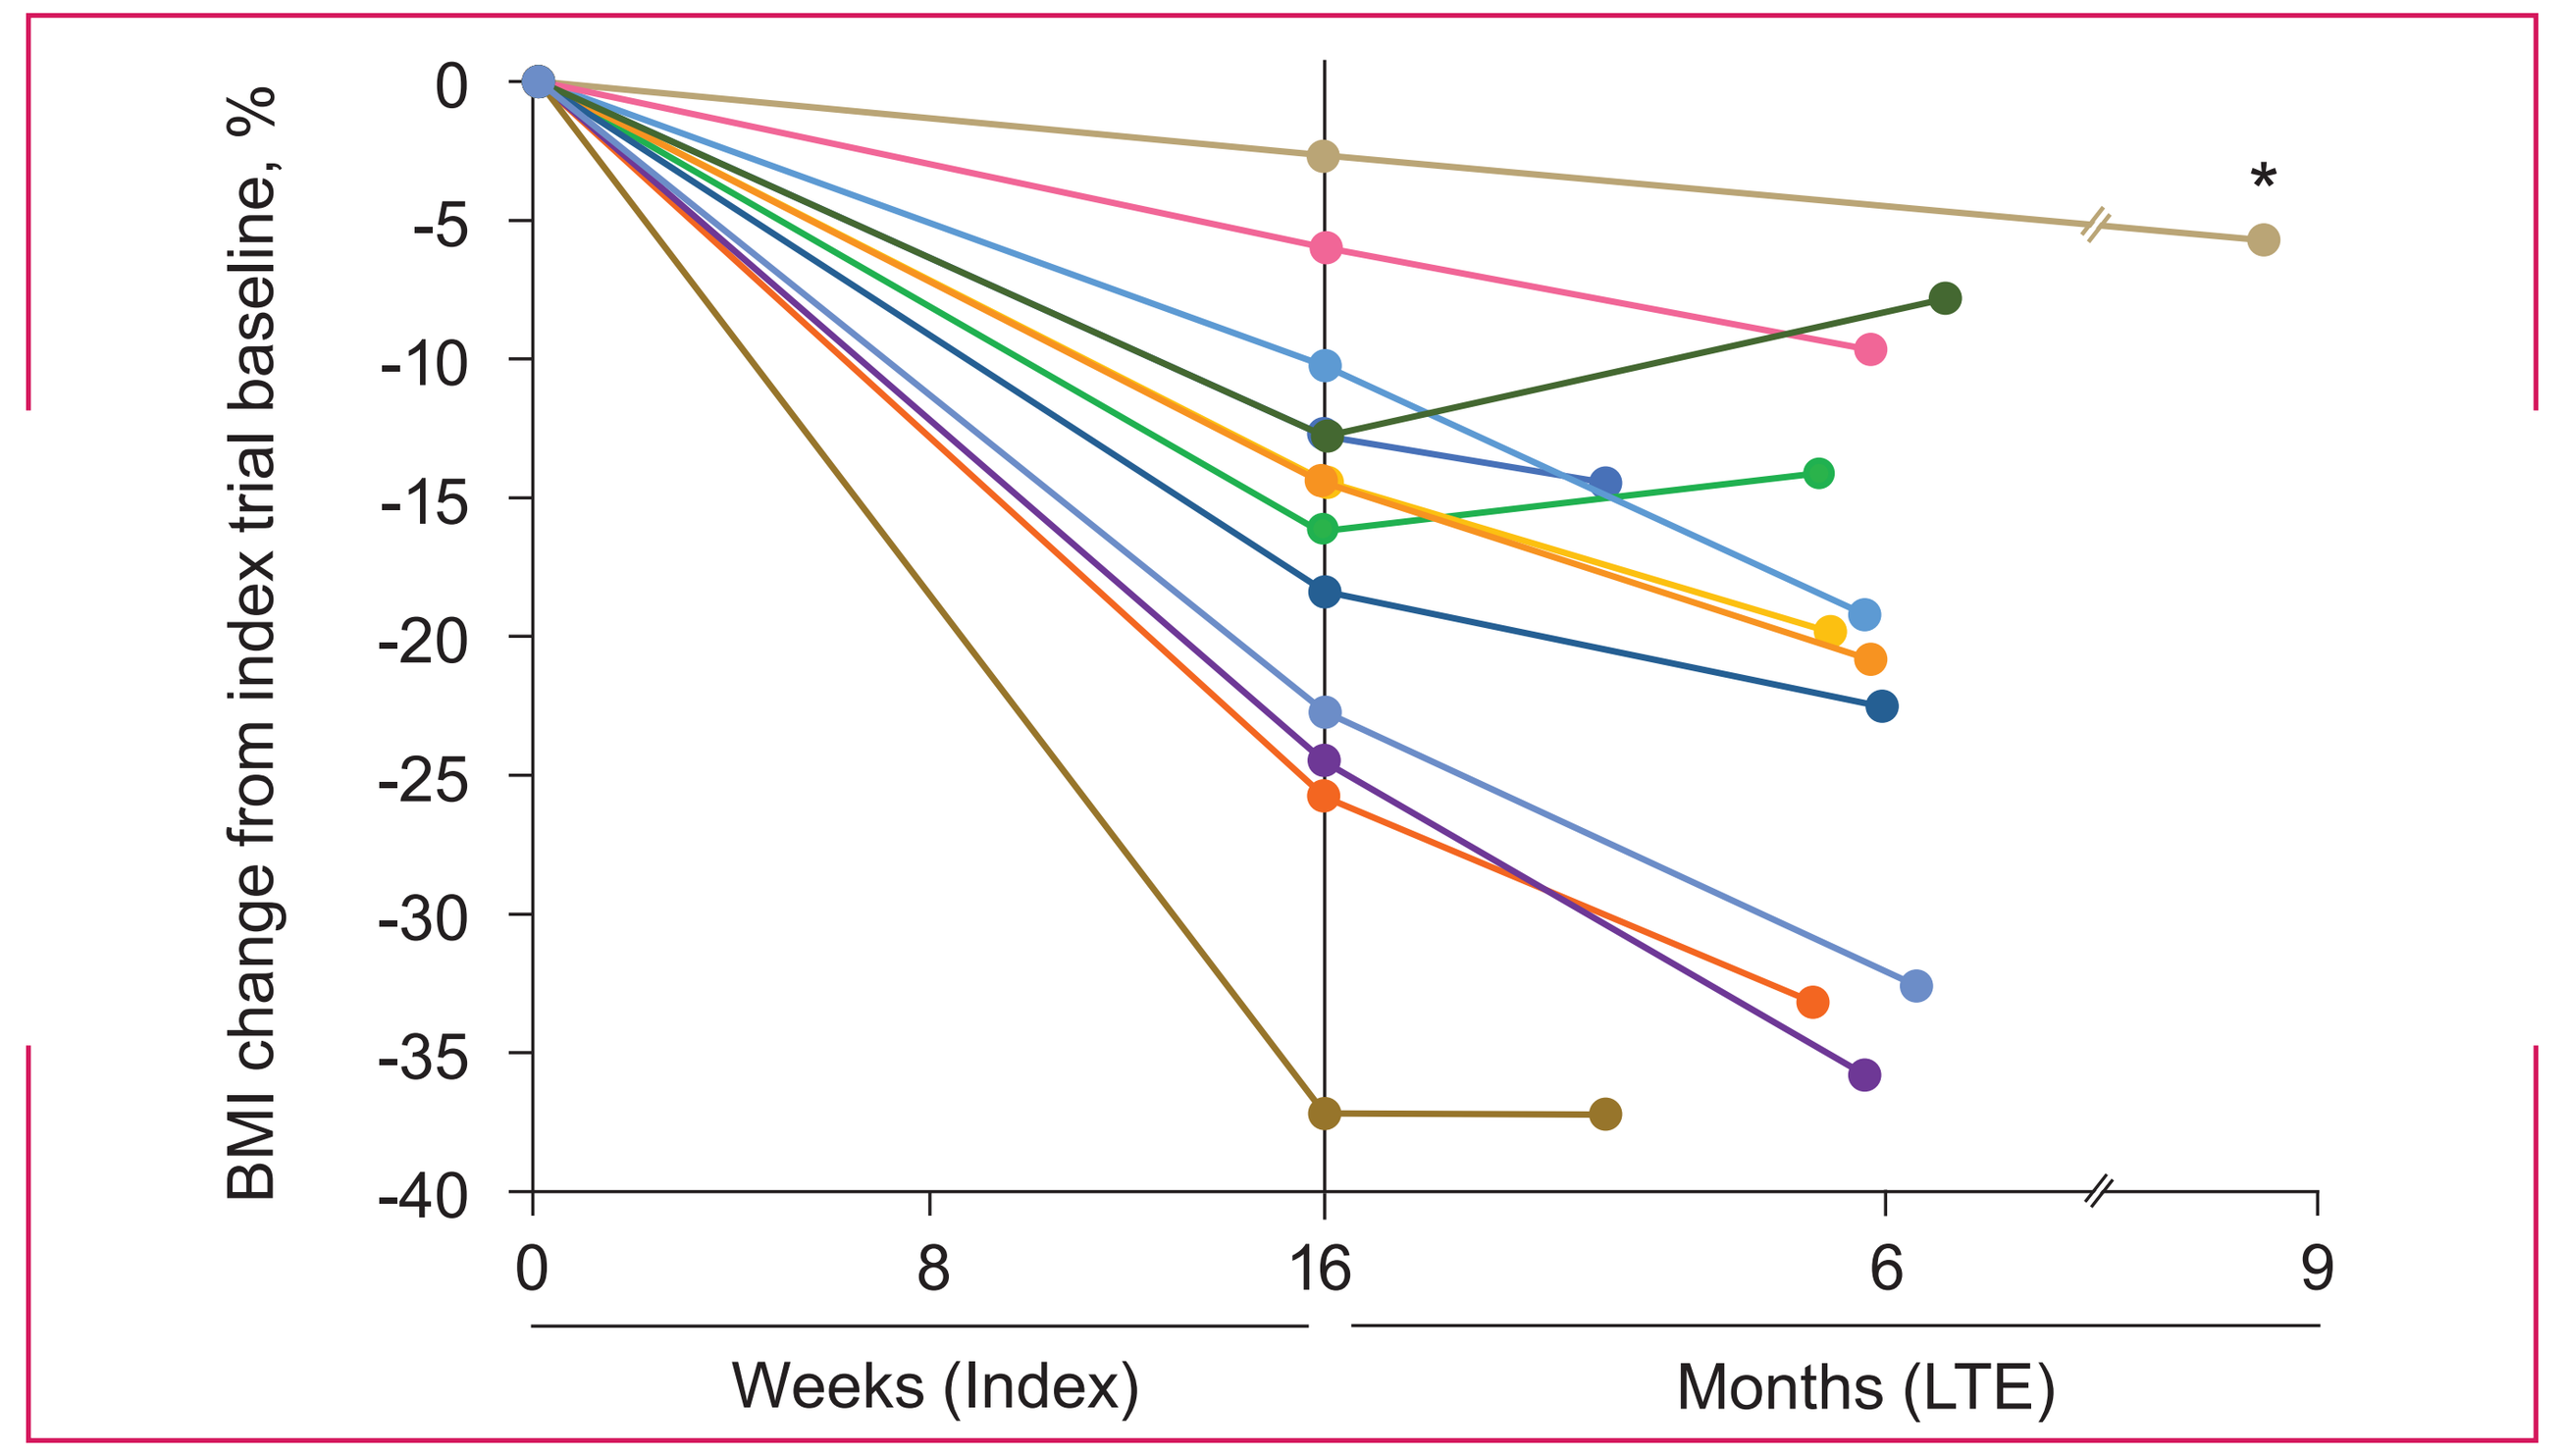
BMI change from index trial baseline, %
0
-5
-10
-15
-20
-25
-30
-35
-40
0
8
16
6
9
*
Weeks (Index)
Months (LTE)

*One patient treated for a hamartoma experienced an increase in BMI during dose escalation in the index trial. This patient's BMI has currently decreased by −5.71% (Month 9); this patient did not have a Month-6 visit. BMI, body mass index; LTE, long-term extension.

- The overall percent change in BMI from baseline at Month 6 was −21.0% (individual percent changes shown in Figure 2)
- At Month 6, 100% (90% confidence interval [CI], 79.4%-100.0%; N=13) of patients achieved ≥5% BMI reduction from index trial baseline and 76.9% (90% CI, 50.4%-93.4%; n/N=10/13) achieved ≥10% reduction in BMI from index trial baseline (Figure 3)

Figure 3. BMI percent change from index trial baseline at Week 16 and Month 6.

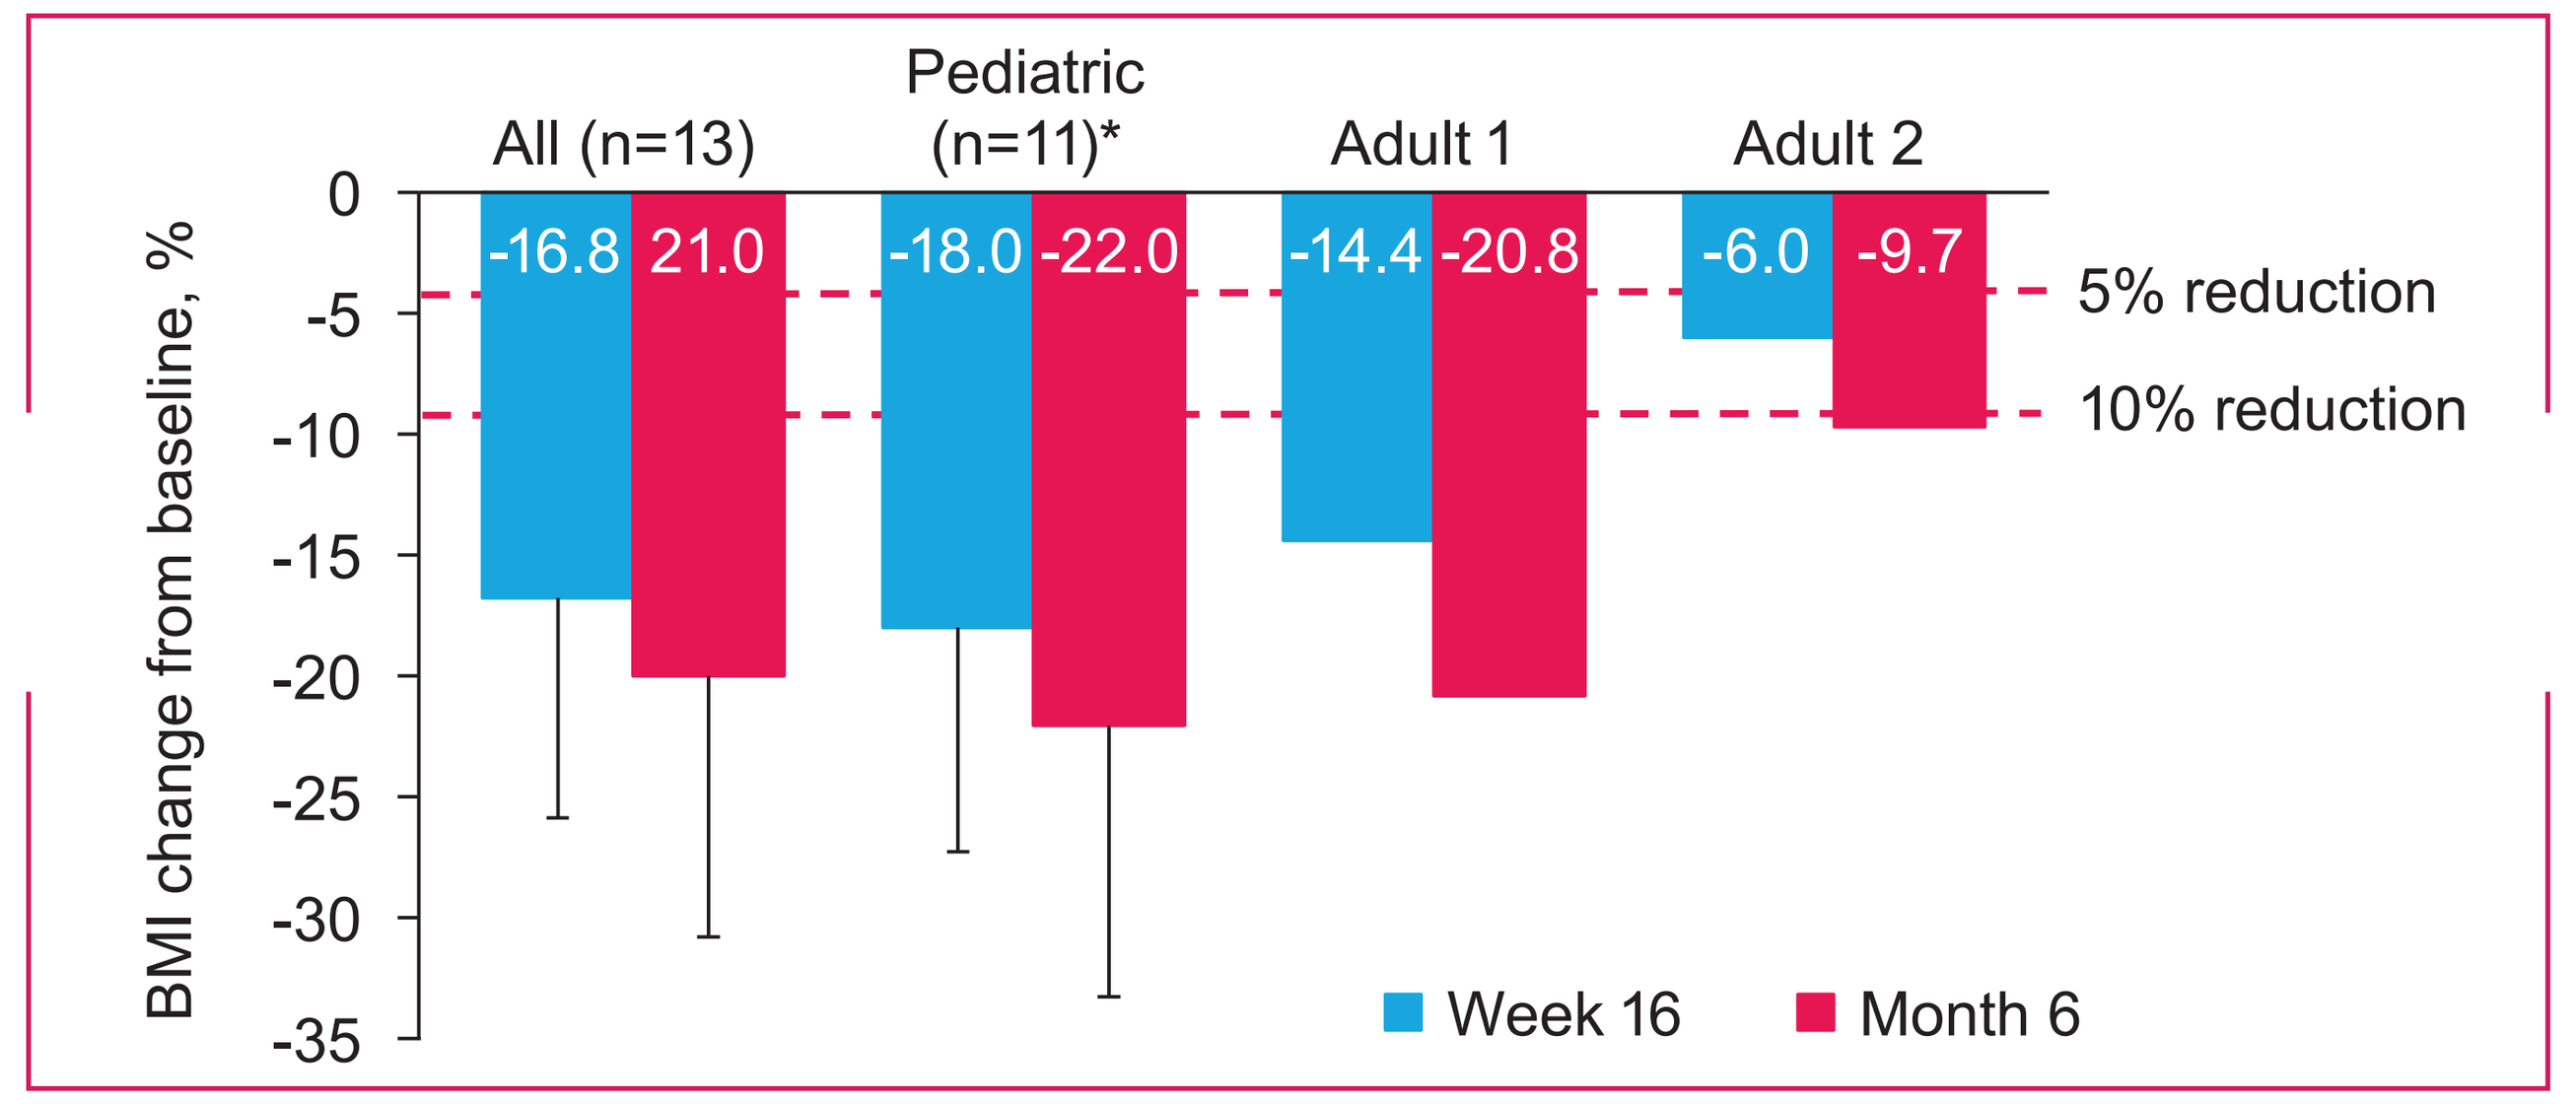
All (n=13)
Pediatric (n=11)*
Adult 1
Adult 2
-16.8
21.0
-18.0
-22.0
-14.4
-20.8
-6.0
-9.7
5% reduction
10% reduction
BMI change from baseline, %
0
-5
-10
-15
-20
-25
-30
-35
Week 16
Month 6

Error bars are the standard deviation. *One pediatric patient did not have a Month-6 visit, and Month-9 data were used for this analysis. BMI, body mass index.

- All patients experienced a decrease in the severity of obesity at Month 6 (Figure 4)
 - Ten of 13 patients (76.9%) improved by ≥1 weight class (based on BMI or BMI percentile); the remaining 3 patients, who had obesity class III at baseline, had reductions of 10 kg/m² in BMI, and 47 and 16 percentage points in %BMI95
 - Four of 11 pediatric patients (36.4%) improved by ≥2 weight classes, and 4 patients moved from different classes of obesity at baseline to the "overweight" or "normal weight" class
- Pediatric patients (n=11) had a decrease in BMI Z score and percent of the BMI 95th percentile at Month 6, with mean (SD) changes of −1.7 (1.1) and −34.3 (15.7), respectively (Figure 5)
- Significant mean decreases of −9.2 kg (−23.9%) and −3.1 kg (−6.3%) were also observed in fat mass and lean muscle mass, respectively, at Week 16 (Figure 6)

Figure 4. Weight class change across individual patients from baseline to Month 6.

BMI, kg/m²	Adults (n=2)		WHO Classification (NIH[5])	Pediatric patients (n=11)											BMI percentile[6]	
≥50	50		Obesity class III (Extreme)									190			≥140%‡	≥95th percentile
≥45 to <50						157	166						158			
≥40 to <45	40								149	144		143	142	141		
≥35 to <40		38*	Obesity class II (Severe[6])	139	124†	133	130				120				≥120% to <140%§	
≥30 to <35		34	Obesity class I	118	113			109	97						≥95% to <120%¶	
≥25 to <30			Overweight					87		91				94	≥85th to <95th percentile	
<25			Normal weight								74				≥5th to <85th percentile	

*Patient reduced dose because of tolerability in the index trial and then titrated back. †Patient had an increase in BMI during dose titration of the index trial; this patient did not have a Month-6 visit and data shown are Month-9 %BMI95. ‡Or BMI ≥40 kg/m² (whichever is lower). §or BMI ≥35 to <40 kg/m² (whichever is lower). ¶or BMI ≥30 to <35 kg/m² (whichever is lower). %BMI95, percent of the BMI 95th percentile; BMI, body mass index; NIH, National Institutes of Health; SD, standard deviation; WHO, World Health Organization.

Figure 5. BMI Z score and %BMI95 change from index trial baseline at Week 16 and Month 6* (n=11).

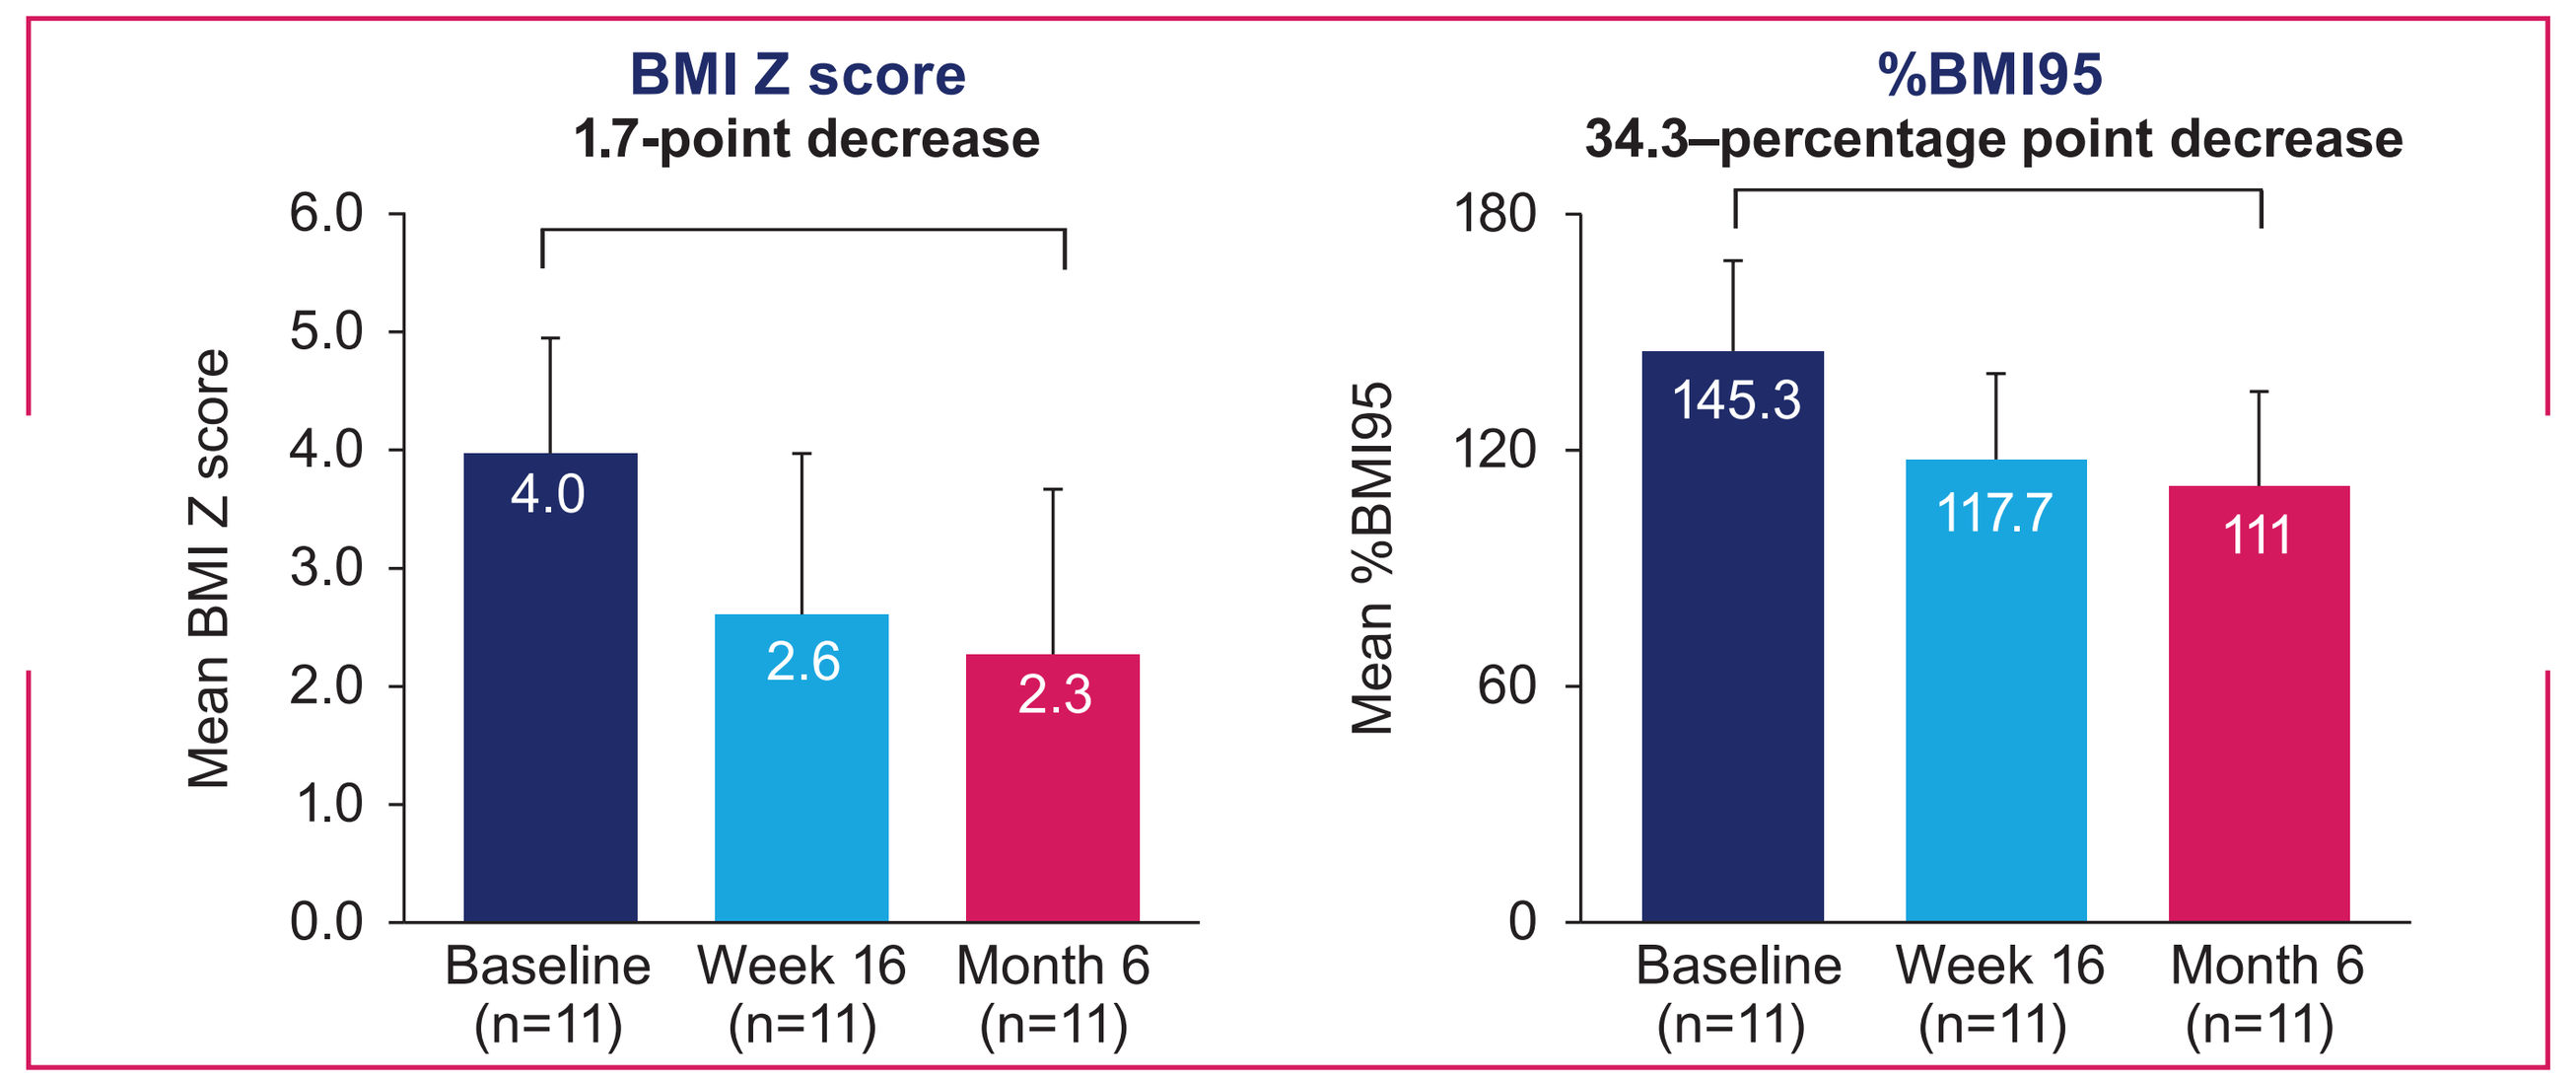
BMI Z score
1.7-point decrease
Mean BMI Z score
6.0
5.0
4.0
3.0
2.0
1.0
0.0
4.0
2.6
2.3
Baseline (n=11)
Week 16 (n=11)
Month 6 (n=11)
%BMI95
34.3–percentage point decrease
Mean %BMI95
180
120
60
0
145.3
117.7
111
Baseline (n=11)
Week 16 (n=11)
Month 6 (n=11)

*One pediatric patient did not have a Month-6 visit, and Month-9 data were used for this analysis. Error bars are the standard deviation. %BMI95, percent of the BMI 95th percentile; BMI, body mass index.

Figure 6. Week-16 body composition in pediatric patients.

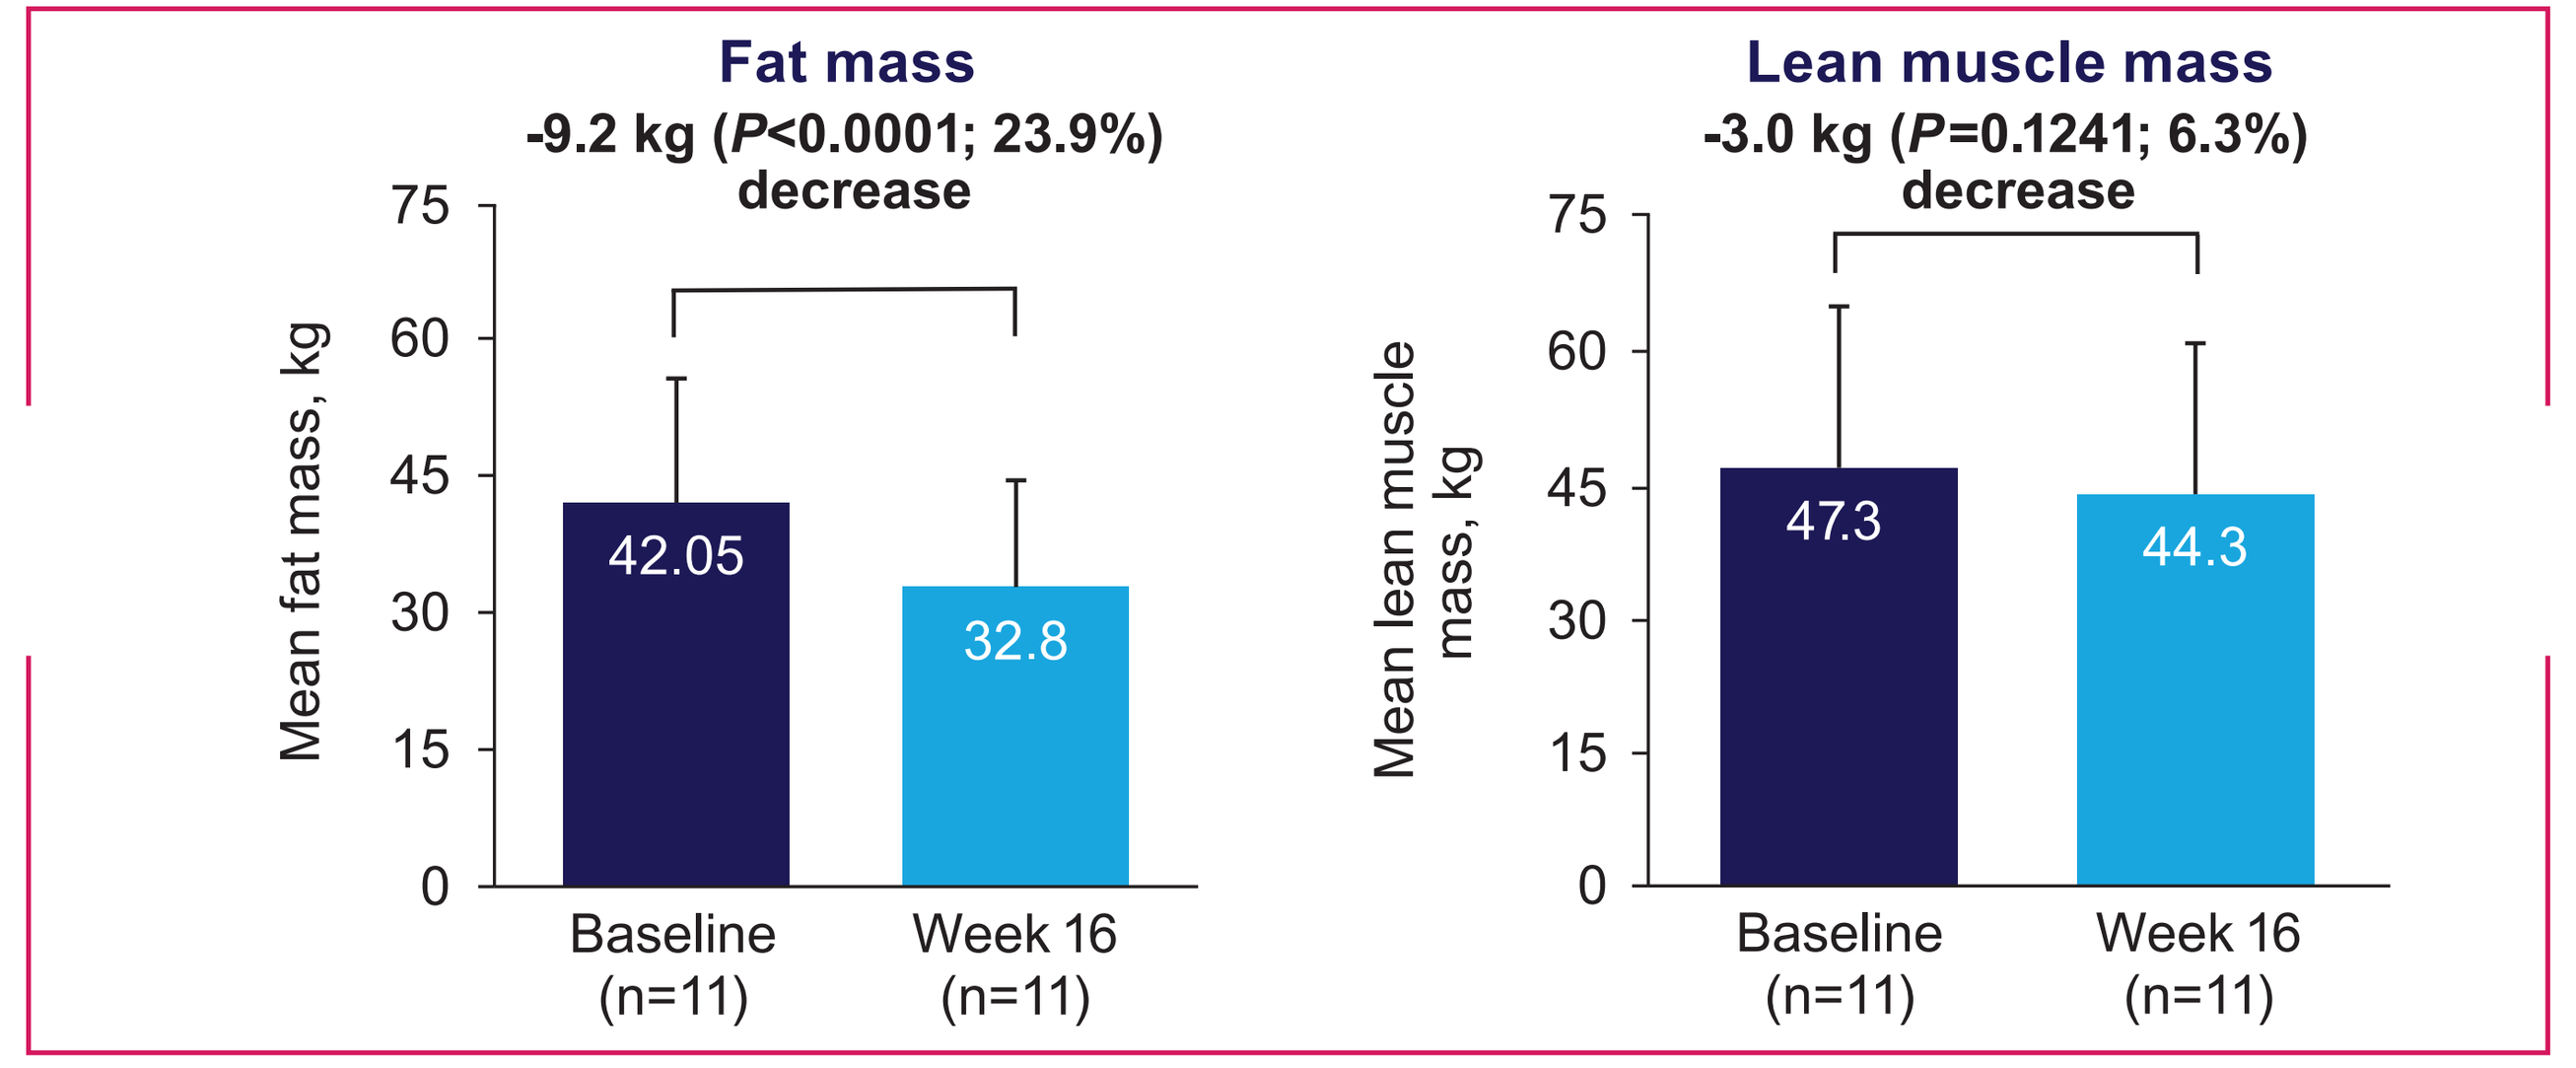
Fat mass
-9.2 kg (P<0.0001; 23.9%) decrease
Mean fat mass, kg
75
60
45
30
15
0
42.05
32.8
Baseline (n=11)
Week 16 (n=11)
Lean muscle mass
-3.0 kg (P=0.1241; 6.3%) decrease
Mean lean muscle mass, kg
75
60
45
30
15
0
47.3
44.3
Baseline (n=11)
Week 16 (n=11)

Error bars are the standard deviation.

Safety outcomes

- AEs from the index and LTE trials were pooled
- AEs of any causality occurred in all patients (n=13)
- No treatment-related AEs led to drug or study discontinuation
- One serious AE reported (*Clostridioides difficile* colitis) was determined not to be related to the study drug
- The most frequent AEs (≥3 patients) were nausea (n=8), skin hyperpigmentation (n=6), vomiting (n=3), injection site pain (n=3), and upper respiratory tract infection (n=3); increase in erections was observed in 4 of 9 male patients
- No new safety concerns were observed in the LTE

Conclusions

- At first LTE follow-up (10 additional weeks beyond the index trial; 26 weeks of total treatment), most patients continue to show progressive improvement in BMI and associated measures with the synthetic analogue setmelanotide
- Most patients (84.6%) achieved ≥1 weight class improvement, and 4 of 11 pediatric patients dropped below the 95th percentile for BMI
- The clinical response to setmelanotide continues to suggest the MC4R pathway plays a critical role in the pathophysiology of HO by replacing a deficit in the endogenous hormone α–melanocyte-stimulating hormone with the synthetic analogue
- Setmelanotide may provide a meaningful therapeutic alternative for a disease that has no approved therapies to date

Acknowledgments: This study was sponsored by Rhythm Pharmaceuticals, Inc. Writing and editorial support for this poster were provided under the direction of the authors by MedThink SciCom and funded by Rhythm Pharmaceuticals, Inc.

Disclosures: MJA, MG, and AHS have received compensation for consulting, advisory board participation, and speaking engagements from Rhythm Pharmaceuticals, Inc. MJA, AHS, MG, JM, and CLR's institution have received research support from Rhythm Pharmaceuticals, Inc. GY, SM, and CS are employees of Rhythm Pharmaceuticals, Inc., and have company awarded RSU and options.

References: **1.** Kim and Choi. *Ann Pediatr Endocrinol Metab.* 2013;18:161-167. **2.** Rose et al. *Obesity (Silver Spring).* 2018;26:1727-1732. **3.** Dimitri. *Front Endocrinol (Lausanne).* 2022;13:846880. **4.** Roth et al. Presented at: ObesityWeek; November 1-4, 2022; San Diego, CA. **5.** National Institutes of Health. *Obes Res.* 1998;6 suppl 2:51S-209S. **6.** Hampl et al. *Pediatrics.* 2023;151:e2022060640.

The Endocrine Society Annual Meeting • June 15-18, 2023 • Chicago, IL