Effect of Setmelanotide on Metabolic Parameters and Vital Signs in a Phase 2 Trial of Patients With Hypothalamic Obesity

M. Jennifer Abuzzahab,[1] Ashley H. Shoemaker,[2] Michael Gottschalk,[3] Jennifer Miller,[4] Guojun Yuan,[5] Sonali Malhotra,[5-7] Cecilia Scimia,[5] Christian L. Roth[8,9]

[1]Pediatric Endocrinology and Diabetes, Children's Minnesota, Saint Paul, MN, USA; [2]Ian Burr Division of Endocrinology and Diabetes, Vanderbilt University Medical Center, Nashville, TN, USA; [3]Pediatric Endocrinology, University of California San Diego/Rady Children's Hospital, San Diego, CA, USA; [4]Pediatric Endocrinology, Department of Pediatrics, University of Florida College of Medicine, Gainesville, FL, USA; [5]Rhythm Pharmaceuticals, Inc., Boston, MA, USA; [6]Massachusetts General Hospital, Boston, MA, USA; [7]Harvard Medical School, Boston, MA, USA; [8]Seattle Children's Research Institute, Seattle, WA, USA; [9]Division of Endocrinology, Department of Pediatrics, University of Washington, Seattle, WA, USA

FRI-082

Presenting Author:
Michael Gottschalk
mgottschalk@rchsd.org

Summary

- **After 16 weeks of treatment with the melanocortin-4 receptor (MC4R) agonist setmelanotide, patients with hypothalamic obesity (HO) completing the trial on treatment demonstrated meaningful reductions in body mass index (BMI) and hunger**
- **Furthermore, most patients experienced reduction in waist circumference with favorable changes in body composition and no adverse changes in metabolic, glycemic, or vital sign parameters; the safety profile was manageable**

Introduction

- HO is a severe form of obesity that results from injury to the hypothalamus, most commonly from tumors, tumor resections, and radiotherapy treatment, which can impair MC4R pathway signaling[1-3]
- Patients with HO experience rapid weight gain and appetite changes, and they are often not responsive to existing therapies for obesity[1]
- Hyperphagia, a pathologic, insatiable hunger, is associated with a severe preoccupation with food and food-seeking behaviors, which can contribute to obesity in HO[1,4,5]
- Treatment with setmelanotide, an MC4R agonist, resulted in meaningful weight loss and hunger reduction in patients with MC4R pathway–associated diseases, including HO[6-8]
 - In a Phase 2 study of patients with HO, 88.9% (16/18; *P*<0.0001) achieved ≥5% reduction in BMI after 16 weeks of setmelanotide treatment
 - Thirteen of 15 patients who completed the trial and were adherent to treatment had ≥10% BMI reduction
 - Eleven out of 12 patients who completed the trial, were adherent to setmelanotide treatment, and completed the daily hunger questionnaires experienced a reduction in their maximal daily hunger score at Week 16, regardless of baseline score

Objective

- To summarize exploratory endpoints, including metabolic, glycemic, and vital sign parameters, and provide a complete therapeutic profile of setmelanotide in patients with HO after 16 weeks of setmelanotide treatment

Methods

Trial design

- Patients aged ≥6 to ≤40 years with obesity, defined as BMI ≥95th percentile for those aged ≥6 to <18 years and BMI ≥35 kg/m² for those aged ≥18 years, resulting from hypothalamic injury were enrolled in a Phase 2 open-label trial of setmelanotide (NCT04725240; Figure 1)
 - Additional key inclusion criteria were diagnosis of craniopharyngioma or nonmalignant brain tumor affecting the hypothalamic region and completion of tumor-related treatment ≥6 months before enrollment
- Patients underwent open-label dose titration to a target therapeutic dose of 3.0 mg of setmelanotide, as tolerated, to be delivered subcutaneously once daily
 - Patients aged ≥6 to <16 years received 1.0 and 2.0 mg of setmelanotide once daily for 2 sequential 2-week periods followed by 3.0 mg of setmelanotide once daily for 12 weeks
 - Patients aged ≥16 years received 2.0 mg of setmelanotide once daily for 2 weeks followed by 3.0 mg of setmelanotide once daily for 14 weeks
 - Patients who meet the primary endpoint of ≥5% reduction in BMI were eligible for enrollment in a long-term extension trial

Figure 1. Trial design.

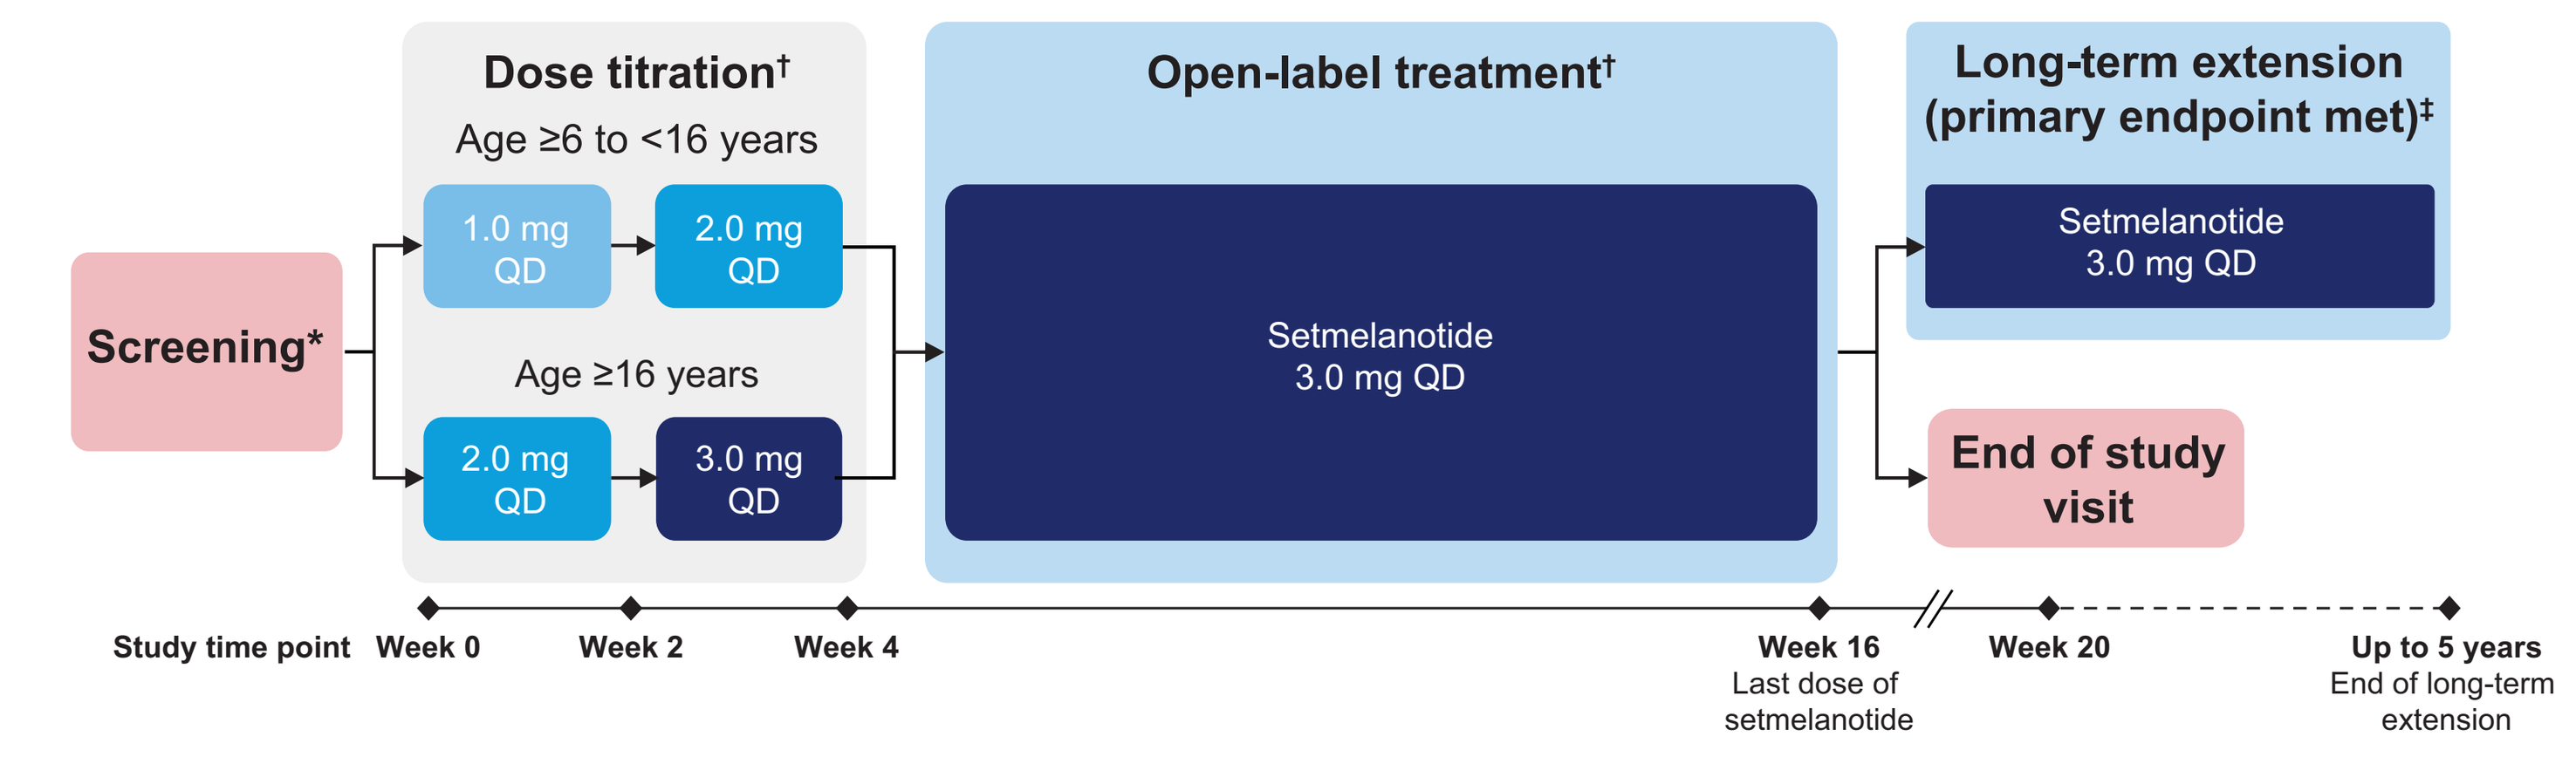
Screening*
Dose titration†
Age ≥6 to <16 years
1.0 mg QD
2.0 mg QD
Age ≥16 years
2.0 mg QD
3.0 mg QD
Open-label treatment†
Setmelanotide 3.0 mg QD
Long-term extension (primary endpoint met)‡
Setmelanotide 3.0 mg QD
End of study visit
Study time point
Week 0
Week 2
Week 4
Week 16
Last dose of setmelanotide
Week 20
Up to 5 years
End of long-term extension

*Diet and/or exercise regimens with or without antiobesity medications were permitted if the regimen and/or dose had been stable for at least 3 months before randomization, the patient had not experienced weight loss ≥2% during the previous 3 months, and the patient intended to continue the regimen throughout treatment. †During the treatment period, including dose titration, patients returned to the clinic every 4 weeks to complete study assessments. ‡Patients meeting the primary endpoint were eligible to enroll in a long-term extension trial of setmelanotide. Patients who did not reach the primary endpoint or elected not to continue setmelanotide returned for an end-of-study visit at Week 20 for a final safety review. BMI, body mass index; QD, once daily.

Outcomes

- The primary endpoint was the proportion of patients with ≥5% reduction in baseline BMI after 16 weeks of setmelanotide treatment
- Exploratory endpoints of change from baseline in waist circumference, body composition as determined by dual-energy X-ray absorptiometry, and glucose parameters, including fasting glucose and glycated hemoglobin (HbA_{1c}), were evaluated
 - Vital signs, including heart rate and blood pressure, were also assessed
- Changes in these parameters at Week 16 from baseline are reported in patients who were adherent to study treatment and were on treatment at Week 16

Results

- Of 18 patients included in the study, 13 (72.2%) were aged ≥6 to <18 years and 5 (27.8%) were aged ≥18 years with the overall mean (SD) age of 15.0 (5.3) years; 13 pediatric and 2 adult patients were adherent to study treatment and on treatment at Week 16, and therefore were included in the analyses (Table 1)
 - One of the adult patients who was included received a subtherapeutic dose because of tolerability
 - Of the 3 adult patients not included, 1 was nonadherent and 2 discontinued because of adverse events
- At baseline, the overall mean (SD) weight and BMI were 102.8 (30.1) kg and 37.95 (6.53) kg/m², respectively

Table 1. Baseline Demographics and Characteristics of Adherent Patients on Treatment at Week 16

	Pediatric patients (n=13)	Adults (n=2)
Age, mean (SD), years	12.3 (3.2)	23.5 (0.7)
Sex		
Male, n (%)	9 (69.2)	1 (50.0)
Female, n (%)	4 (30.8)	1 (50.0)
Weight, mean (SD), kg	96.0 (30.9)	129.3 (29.5)
Fat mass, mean (SD), kg	44.2 (14.2)*	61.5 (20.4)*
Lean muscle mass, mean (SD), kg	48.3 (17.2)*	61.1 (3.7)*
BMI, mean (SD), kg/m²	36.2 (6.2)	44.0 (9.0)
BMI Z score, mean (SD)†	3.94 (0.91)	—
BMI percent of 95th percentile, mean (SD)	144.7 (21.4)	—
Waist circumference, mean (SD), cm	110.0 (16.5)	130.9 (16.5)
Fasting glucose, mean (SD)		
mmol/L	4.6 (0.7)	4.8 (0.5)
mg/dL	83.2 (12.1)	86.4 (6.2)
HbA_{1c}, mean (SD), %	5.3 (0.3)	5.8 (0.4)
Heart rate, mean (SD), beats/minute	88.6 (10.4)	85.0 (2.4)
Systolic blood pressure, mean (SD), mm Hg	109.0 (6.4)	120.5 (13.4)
Diastolic blood pressure, mean (SD), mm Hg	63.3 (8.9)	84.5 (0.2)

*Body composition for fat mass and lean muscle mass was measured using dual-energy X-ray absorptiometry. †The World Health Organization 2007 reference standard growth chart was used to calculate individual BMI Z scores. BMI, body mass index; HbA_{1c}, glycated hemoglobin; SD, standard deviation.

Weight-related outcomes

- Of the 13 pediatric patients, 12 achieved ≥0.2-point BMI Z score reduction from baseline (92.3%; *P*<0.0001); mean (SD) percent change in BMI percent of 95th percentile was −18.5% (8.7%; n=13) (Table 2)
- In adult patients, 2 of 2 achieved ≥5% BMI reduction from baseline and 1 of 2 achieved ≥10% reduction; mean (SD) BMI percent change was −10.2% (5.9%; n=2) (Table 2)
- In pediatric and adult patients, the mean (SD) percent change in waist circumference was −11.1% (5.6%) and −8.0% (0.5%), respectively
- Weight loss was predominately due to reduction in total fat mass rather than loss of lean muscle mass
 - In pediatric patients, mean (SD) changes in lean muscle and fat mass were −3.6 (5.7) kg and −9.2 (3.6) kg, respectively
 - In adult patients, mean (SD) changes in lean muscle and fat mass were −5.2 (3.8) kg and −5.4 (1.6) kg, respectively

Table 2. Change in Weight-Related Measures in Adherent Patients on Treatment at Week 16

	Pediatric patients (n=13)		Adults (n=2)	
Mean (SD)	Change from baseline at Week 16	Percent change from baseline at Week 16	Change from baseline at Week 16	Percent change from baseline at Week 16
Weight, kg	−14.1 (8.4)*	−15.1 (9.2)*	−14.1 (10.7)	−10.2 (5.9)
Fat mass, kg	−9.2 (3.6)*,†	−22.8 (13.0)*	−5.4 (1.6)†	−8.8 (0.4)*
Lean muscle mass, kg	−3.6 (5.7)*,†	−7.1 (12.2)	−5.2 (3.8)†	−8.3 (5.7)
BMI, kg/m²	−6.3 (2.8)*	−17.6 (8.8)*	−4.8 (3.5)	−10.2 (5.9)
BMI Z score‡,§	−1.28 (1.04)	—	—	—
BMI percent of 95th percentile‡	−27.0 (12.1)	—	—	—
Waist circumference, cm	−12.6 (6.3)*	−11.1 (5.6)*	−10.4 (0.7)*	−8.0 (0.5)*

**P*<0.05. †Body composition for fat mass and lean muscle mass was measured using dual-energy X-ray absorptiometry. ‡*P*-value not evaluated. §The World Health Organization 2007 reference standard growth chart was used to calculate individual BMI Z scores. BMI, body mass index; SD, standard deviation.

Glucose parameters and vital signs

- Mean fasting glucose and HbA_{1c} were within normal ranges at baseline, and no significant changes were observed at Week 16 (Table 3)
 - At Week 16 of setmelanotide treatment, in pediatric and adult patients, the mean (SD) change in fasting glucose was 0.25 (0.67) mmol/L (4.5 [12.1] mg/dL) (n=10) and −0.22 (not applicable [NA]) mmol/L (−4.0 mg/dL [NA]) (n=1), respectively
 - In pediatric and adult patients, the mean (SD) change in HbA_{1c} was −0.02 (0.25) (n=9) and −0.20 (NA) (n=1), respectively
- Vital signs, including heart rate and blood pressure, were also within reference ranges at baseline and remained within those ranges during the 16-week study duration with no significant changes
 - In pediatric patients (n=13), the mean (SD) changes in heart rate, systolic blood pressure, and diastolic blood pressure were −4.0 (10.6) beats/minute, −2.1 (8.1) mm Hg, and 1.6 (7.1) mm Hg, respectively, at Week 16
 - In adult patients (n=2), the mean (SD) changes in heart rate, systolic blood pressure, and diastolic blood pressure were −16.2 (26.6) beats/minute, 11.3 (15.1) mm Hg, and −8.3 (1.9) mm Hg, respectively, at Week 16

Table 3. Change From Baseline in Glucose and Vital Sign Measures for Adherent Patients on Treatment at Week 16

	Pediatric patients (n=13)		Adults (n=2)	
Mean (SD)	Change from baseline at Week 16	Percent change from baseline at Week 16	Change from baseline at Week 16	Percent change from baseline at Week 16
Fasting glucose				
mmol/L	0.3 (0.7)*	8.43 (21.6)†	−0.2 (NA)‡	−5.0 (NA)
mg/dL	4.5 (12.1)*,†	—	−4.0 (NA)‡	—
HbA_{1c}, %	0.0 (0.3)†,§	−0.5 (4.7)†,§	−0.2 (NA)‡	−3.3 (NA)‡
Heart rate, beats/minute	−4.0 (10.6)†	−4.2 (11.9)†	−16.2 (26.6)†	−18.6 (30.8)†
Systolic blood pressure, mm Hg	−2.1 (8.1)†	−1.9 (7.7)†	11.3 (15.1)†	10.2 (13.7)†
Diastolic blood pressure, mm Hg	1.6 (7.1)†	3.0 (11.4)†	−8.3 (1.9)†	−9.9 (2.3)†

*n=10. †*P*>0.05. ‡n=1. §n=9. HbA_{1c}, glycated hemoglobin; NA, not applicable; SD, standard deviation.

Safety outcomes

- Treatment-emergent adverse events (AEs) occurred in all patients in the safety population (N=18; Table 4)
 - The most frequent AEs included nausea (61.1%; n=11), vomiting (33.3%; n=6), skin hyperpigmentation (33.3%; n=6), and diarrhea (22.2%; n=4)

Table 4. AEs of All Patients Entering the Trial

	All patients (N=18), n (%)	Adherent patients on treatment at Week 16 (n=15), n (%)
Overall AEs	18 (100.0)	15 (100.0)
AEs occurring in ≥10% of the overall population*		
Nausea	11 (61.1)	9 (60.0)
Vomiting	6 (33.3)	5 (33.3)
Skin hyperpigmentation	6 (33.3)	5 (33.3)
COVID-19	4 (22.2)	3 (20.0)
Diarrhea	4 (22.2)	3 (20.0)
Abdominal pain	3 (16.7)	2 (13.3)
Fatigue	3 (16.7)	3 (20.0)
Injection site pain	3 (16.7)	3 (20.0)
Erection increased	3 (16.7)†	3 (33.3)‡
Injection site pruritus	2 (11.1)	2 (13.3)
Pain	2 (11.1)	2 (13.3)
Pyrexia	2 (11.1)	1 (6.7)
Upper respiratory tract infection	2 (11.1)	2 (13.3)
Headache	2 (11.1)	2 (13.3)
Contusion	2 (11.1)	1 (6.7)
Back pain	2 (11.1)	2 (13.3)
Hypertransaminasemia	2 (11.1)	1 (6.7)
Treatment-related AEs	15 (83.3)	13 (86.7)
Serious AEs	1 (5.6)	1 (6.7)
Serious treatment-related AEs	0 (0)	0
AEs leading to study discontinuation	2 (11.1)	0
AEs leading to death	0 (0)	0

*AEs are coded using MedDRA Version 24.0. Patients are counted once for each preferred term. Percentages are calculated using the total number of patients unless otherwise noted. †Denominator of the male population (n=11). ‡Denominator of the male population (n=9). AE, adverse event.

- There was 1 serious AE of *Clostridium difficile* colitis, determined not related to treatment
- Two patients in the overall population entering the trial (11.1%) discontinued because of AEs of hyperpigmentation (n=1) and increased levels of aminotransferase (n=1)

Conclusions

- In a heterogeneous population of patients with HO secondary to treatment of hypothalamic tumors, 16 weeks of setmelanotide treatment was previously shown to reduce weight and BMI
- Here we show that 16 weeks of setmelanotide treatment resulted in favorable changes in body composition with no effect on glucose parameters or vital signs, which continued to remain within normal ranges
- The safety profile of setmelanotide was consistent with prior clinical experience; the most common AE was nausea, which typically occurred during the first weeks of setmelanotide treatment, was transient and not severe, and did not lead to discontinuation
- Efficacy and safety of setmelanotide in patients with HO will continue to be evaluated in a larger Phase 3 clinical trial

Acknowledgments: This study was sponsored by Rhythm Pharmaceuticals, Inc. Writing and editorial support for this poster were provided under the direction of the authors by MedThink SciCom and funded by Rhythm Pharmaceuticals, Inc.

Disclosures: MJA has received compensation for consulting, advisory board participation, and speaking engagements from Rhythm Pharmaceuticals, Inc.; Pfizer; Endo Pharmaceuticals; and ConSynance Therapeutics. MJA's institution has received research support from Rhythm Pharmaceuticals, Inc.; Ascendis Pharma; Novo Nordisk; Levo Therapeutics; Lumos Pharma; Saniona; and Soleno Therapeutics. AHS has received payments from Rhythm Pharmaceuticals, Inc., and Saniona for advisory boards and lectures. AHS's institution has received research support from Rhythm Pharmaceuticals, Inc. MG has received compensation for consulting and participation on advisory boards from Rhythm Pharmaceuticals, Inc. MG's institution has received funding for clinical trials from Rhythm Pharmaceuticals, Inc. JM has received study funding from Rhythm Pharmaceuticals, Inc. GY is an employee of Rhythm Pharmaceuticals, Inc., and has company-awarded RSU and options. SM is an employee and stockholder in Rhythm Pharmaceuticals, Inc. CS is an employee of Rhythm Pharmaceuticals, Inc., and has company-awarded RSU and options. CLR's institution has received research support from Rhythm Pharmaceuticals, Inc.

References: **1.** Abuzzahab et al. *Horm Res Paediatr*. 2019;91:128-136. **2.** Erfurth. *Neuroendocrinology*. 2020;110:767-779. **3.** Rose et al. *Obesity (Silver Spring)*. 2018;26:1727-1732. **4.** Hampl et al. *Pediatrics*. 2023;151:e2022060640. **5.** Ludwig and Friedman. *JAMA*. 2014;311:2167-2168. **6.** Roth et al. Poster presented at: ObesityWeek®; November 1-4, 2022; San Diego, CA. **7.** Clément et al. *Lancet Diabetes Endocrinol*. 2020;8:960-970. **8.** Haqq et al. *Lancet Diabetes Endocrinol*. 2022;10:859-868.

The Endocrine Society Annual Meeting • June 15-18, 2023 • Chicago, IL